SUN-253 ENDO 2024

Skeletal Effects among Pre and Postmenopausal Women with Hypoparathyroidism (HypoPT): Data from the Canadian National Hypoparathyroidism Registry (CNHR)

Dalal S. Ali[1], Aliya A. Khan[1,2], H AbuAlrob[1,2], M Ovize[6], Zayd Al Kassem[1], A Almoulia[2] H. Affifi, R Abu Fardeh[1,2], A Azharruddin[1,2], D Bole[2], M Braga[1,2], P Chandra[3], A Cheng[4], R Cheung[5],, N Firdous[1,2], H Juta[2], S Khan[2,4], T Khan[5], J Malhem[2], H Malik[1,2], A Millar[4], E Morgante[2], F Naveed[2], H Niazi[2], T Paul[5], A Prebtani[1], Z Punthakee[1], J Shaban[5], R Shah[1], M Shrayyef[4], M Shaikh[1,2], C Tagra[1,2], S Teschke[1], I Treki[5], R Tauqir[1,2], S Van Uum[5], R Wilson[5], JEM Young[1], B Weiss[6]

McMaster University[1], Bone Research & Education Centre[2], Brampton Hospital[3], University of Toronto[4], Western University[5], Amolyt Pharma, Ecully, France[6]

INTRODUCTION

Chronic hypoparathyroidism (cHP) is a rare endocrine disorder associated with decreased bone turnover and abnormalities in bone density, microarchitecture, and possibly strength. Current international guidelines do not recommend a systematic evaluation of bone health in cHP patients. Our study aimed to systematically assess bone health in cHP patients and, specifically, in postmenopausal women demonstrating progressive reduction in bone mineral density (BMD).

PATIENTS AND METHODS

This prospective study enrolled 101 adult cHP patients from the Canadian National Hypoparathyroidism Registry (CNHR). 83 (82%) were female, and 18 (18%) were male. There were 35 (42%) premenopausal females and 48 (58%) postmenopausal females (PMF). Only PMF were on antiresorptive therapy. Seven (7/8) were on bisphosphonate therapy (i.e., alendronate), and 3 females were on denosumab.

Demographic and clinical characteristics, history of the disease and bone fractures, biochemistry, and serum bone biomarkers were assessed at baseline (Table 1). Bone health evaluation included assessments of fragility fractures, BMD, and trabecular bone score (TBS) by dual-energy X-ray absorptiometry at lumbar spine, femoral neck, total hip, and 1/3 radial sites, and bone markers.

	Study population (n=101)	Men (n=18)	Women (n=83)	
			Premenopausal (n=35)	Postmenopausal (n=48)
Age, years (SD)	52.8 ± 16.0	50 ± 19	39.4 ± 8.5	63.7 ± 10.0
BMI, kg/m² (SD)	28.8 ± 7.9	30 ± 9	28.1 ± 8.1	28.8 ± 7.3
Duration of cHP, years (SD)	11.2 ± 8.6	15 ± 9	8.1 ± 6.8	11.9 ± 8.8
Postsurgical etiology	75	11	24	40
Non-surgical etiology	26	7	11	8

Table 1. Baseline characteristics of the study population and by subgroups (N=101)

RESULTS

Low BMD was confirmed by T-score <-1.0 and >-2.5 at any site in postmenopausal women and men ≥50 years, and by Z-score ≤-2 at any site in premenopausal women and men <50 years. Overall, 12 patients (11.9%, n=101) showed low BMD, all of whom were postmenopausal women. The prevalence of low BMD in postmenopausal women was 25.0% (n=12/48). Low BMD was not present in premenopausal women or men in this cohort.

In total, 15 postmenopausal women (31.3%, n=48) and 2 men ≥50 years (22.2%, n=9) showed osteoporotic T-scores (≤-2.5) at any site. Osteoporosis was more frequent at the lumbar spine (10.4%, n=5/48) and 1/3 radius (12.5%, n=6/48) among postmenopausal women, whereas total hip (11.1%, n=1/9) and 1/3 radius (11.1%, n=1/9) were osteoporotic sites in men ≥50 years. The distribution of patients with low BMD and osteoporosis is presented in Figure 1.

Overall, 11.9% patients (n=12/101) reported a history of fragility fractures, most of whom were postmenopausal women (83.3%, n=10/12). **35% of PMF and 33.3% of men over 50 yrs have osteoporosis by BMD or fragility fracture.**

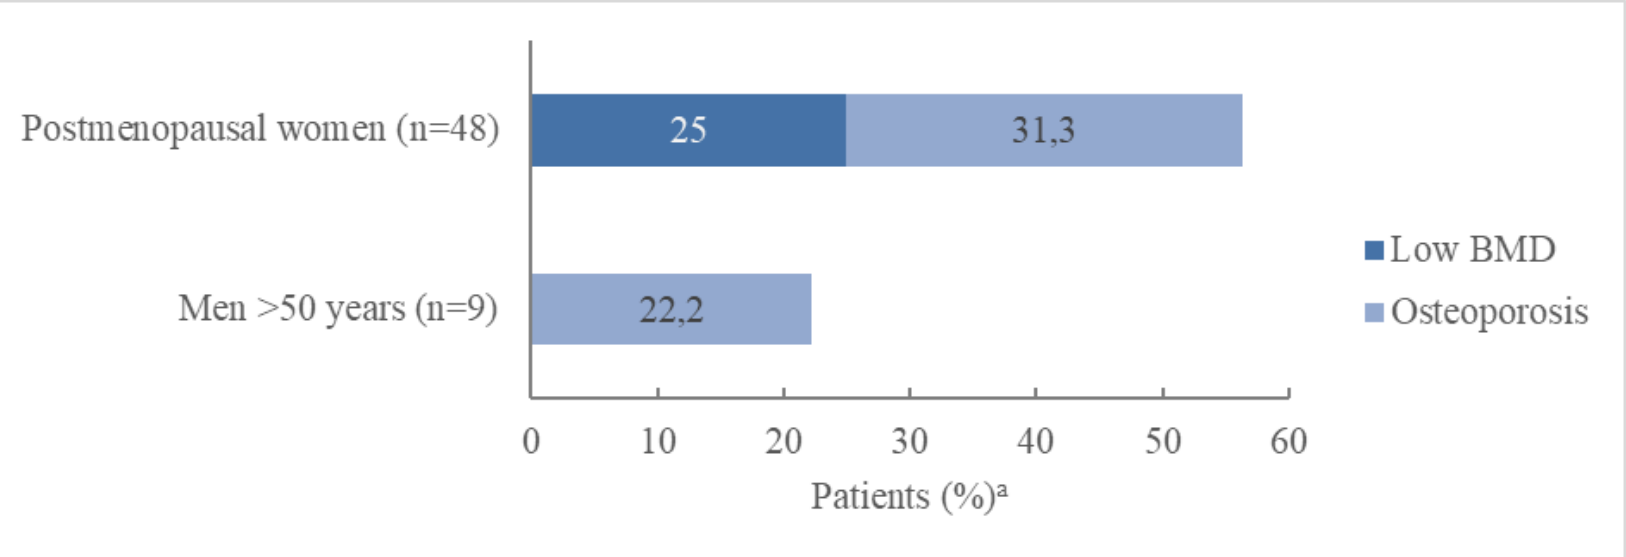

Postmenopausal women (n=48)
Men >50 years (n=9)
25
31,3
22,2
Low BMD
Osteoporosis
0 10 20 30 40 50 60
Patients (%)a

Figure 1. Distribution of postmenopausal women and men ≥50 years with low BMD and osteoporosis

CONCLUSIONS

The results of this prospective cohort study demonstrated significant bone fragility in postmenopausal cHP women of the CNHR, as shown by a higher-than-expected proportion of low BMD, osteoporosis and fractures. These findings suggest that contrary to common belief and despite usually higher BMD, a close follow-up of bone health is necessary in postmenopausal cHP women and men over the age of 50.

REFERENCES

1. Khan AA, et al. Canadian national hypoparathyroidism registry: an overview of hypoparathyroidism in Canada. Endocrine. 2021 May;72(2):553-561. doi: 10.1007/s12020-021-02629-w. Epub 2021 Mar 2. PMID: 33655415.
2. Khan AA, et al. Evaluation and Management of Hypoparathyroidism Summary Statement and Guidelines from the Second International Workshop. J Bone Miner Res. 2022 Dec;37(12):2568-2585. doi: 10.1002/jbmr.4691. Epub 2022 Nov 14. PMID: 36054621.

DISCLOSURES

This study has been funded by Amolyt Pharma.